COALITION AGAINST MAJOR DISEASES: DEVELOPMENT OF THERAPEUTIC AREA-SPECIFIC DATA STANDARDS FOR ALZHEIMER'S AND PARKINSON'S DISEASES

Jon Neville[1], Steve Kopko[2], Bob Stafford[1], Andreas Jeromin[3], Jerry Novak[4], Patricia E. Cole[5], Mark Austin[6], Klaus Romero[1], Joanne Odenkirchen[7], Bess LeRoy[1], Emily Hartley[1], Susan De Santi[8], Steve Broadbent[1], Enrique Avilés[1], Diane Stephenson[1]

(1) Critical Path Institute (2) Clinical Data Interchange Standards Consortium (3) Atlantic Biomarkers, LLC (4) Janssen R&D (5) Takeda Pharmaceuticals (6) IXICO (7) National Institute for Neurological Disorders and Stroke (8) Piramal Life Sciences

Background and Objectives

Critical Path Institute (C-Path) has played a leadership role in both consensus data standards and precompetitive data sharing for multiple disease areas including Alzheimer's disease (AD) and Parkinson's disease (PD). Both of these factors are key to success of clinical trials in the future. Working with the Clinical Data Interchange Standards Consortium (CDISC), C-Path has successfully developed consensus data standards for AD and PD, and a version 2.0 of the AD CDISC standards has been recently completed aimed at biomarkers and early stages of the AD spectrum. These therapeutic area specific standards represent the preferred format by regulatory agencies for submitting new drug applications. Importantly, CDISC standards will be required by FDA for regulatory submission as early as FY 2017. Thus, these standards serve two main purposes: integration of existing data and the prospective collection of clinical trial data.

Methods

A coalition of industry members, regulatory agencies, academic experts, government agencies and patient groups collectively developed data standards in partnership with the Clinical Data Interchange Standards Consortium (CDISC). With input from clinical subject matter experts (SMEs), NINDS (for PD) and ADNI (for AD), working groups of data modelers mapped clinical concepts relevant to AD and PD to the CDISC Study Data Tabulation Model (SDTM) and developed controlled terminology to support the construction of standardized databases for research and regulatory submission in AD and PD.

Concepts covered by the Alzheimer's CDISC User Guide
ApoE Genotype
Family History of AD
Volumetric MRI
PET, PET/CT (FDG, Florbetapir, PiB)
CSF Biomarkers and Sampling
Outcome Assessment Scales
ADAS-COG
MMSE
CDR
AVLT
FAQ
Modified Hachinski Ischemic Scale
DAD
ADCS-ADL MCI
NPI
CGI
GDS
Table 1: Concepts covered by the CDISC AD User Guide. Note that additional assessment scales relevant to AD may be available at www.cdisc.org/ft-and-qt

Concepts covered by the Parkinson's CDISC User Guide
MRI
PET-SPECT
Deep Brain Stimulation
Neuropathology-Microscopic Findings
Family History of PD
Biospecimens (Tissue sample handing/tracking)
Outcome Assessment Scales
UPDRS
MDS-UPDRS
Table 2: Concepts covered by the CDISC PD User Guide. Note that additional assessment scales relevant to PD may be available at www.cdisc.org/ft-and-qt

Results

CDISC therapeutic-area data standards implementation guides were developed for AD and PD in collaboration with CDISC as supplements to the CDISC SDTM, a standard recognized by FDA. The AD user guide represents the first ever therapeutic area CDISC standard developed. The remapping of legacy clinical trial data to the AD standard played a critical role in developing an integrated database of legacy clinical trials, which in turn was a key foundation for the development of the first-ever regulatory-endorsed clinical trial simulation tool. Concepts covered by the AD CDISC user guide include CSF biomarkers, ApoE genotype, volumetric MRI, amyloid PET imaging, and more than 10 clinical outcome assessment scales, including ADAS-Cog, MMSE and CDR. Concepts covered by the PD CDISC user guide include MRI, PET-SPECT, deep brain stimulation, neuropathology, and two clinical outcomes assessments scales: UPDRS and MDS-UPDRS.

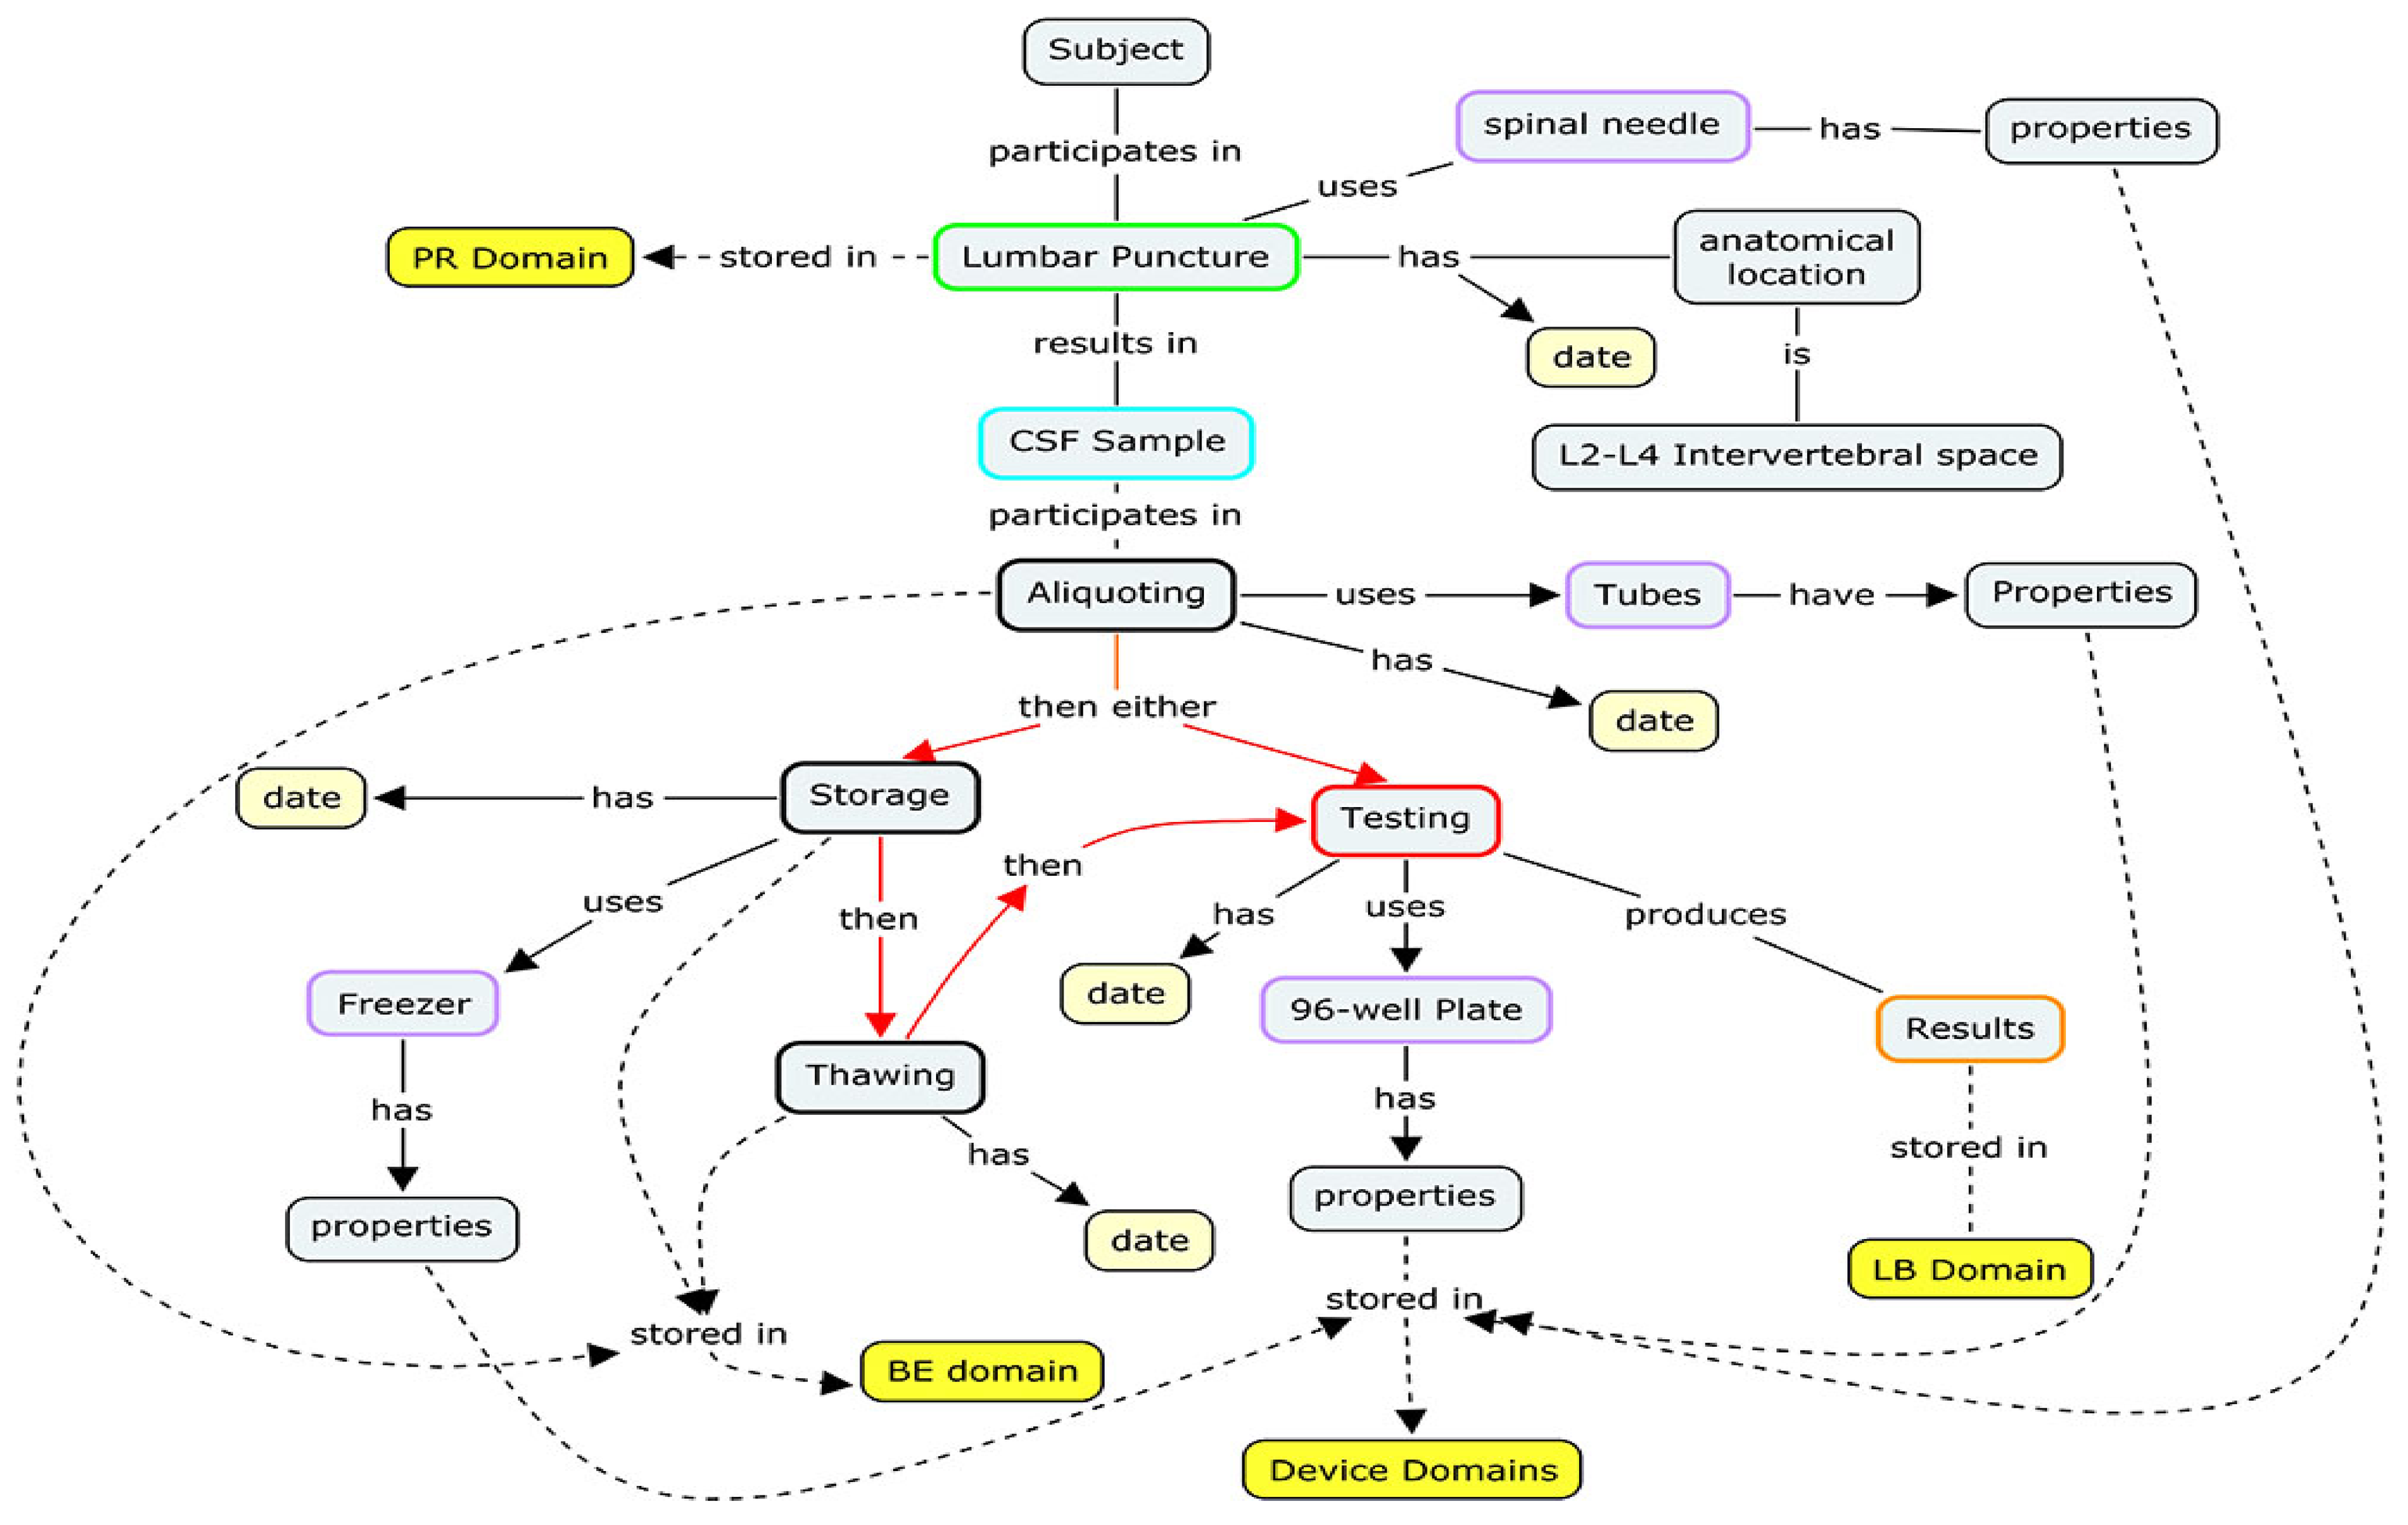
Subject
participates in
spinal needle
has
properties
uses
PR Domain
stored in
Lumbar Puncture
has
anatomical location
date
is
L2-L4 Intervertebral space
results in
CSF Sample
participates in
Aliquoting
uses
Tubes
have
Properties
has
date
then either
date
has
Storage
Testing
then
uses
then
has
uses
produces
Freezer
date
96-well Plate
Results
Thawing
has
has
has
stored in
properties
date
properties
LB Domain
stored in
BE domain
stored in
Device Domains

Panel A: Concept map of CSF sampling and biomarkers

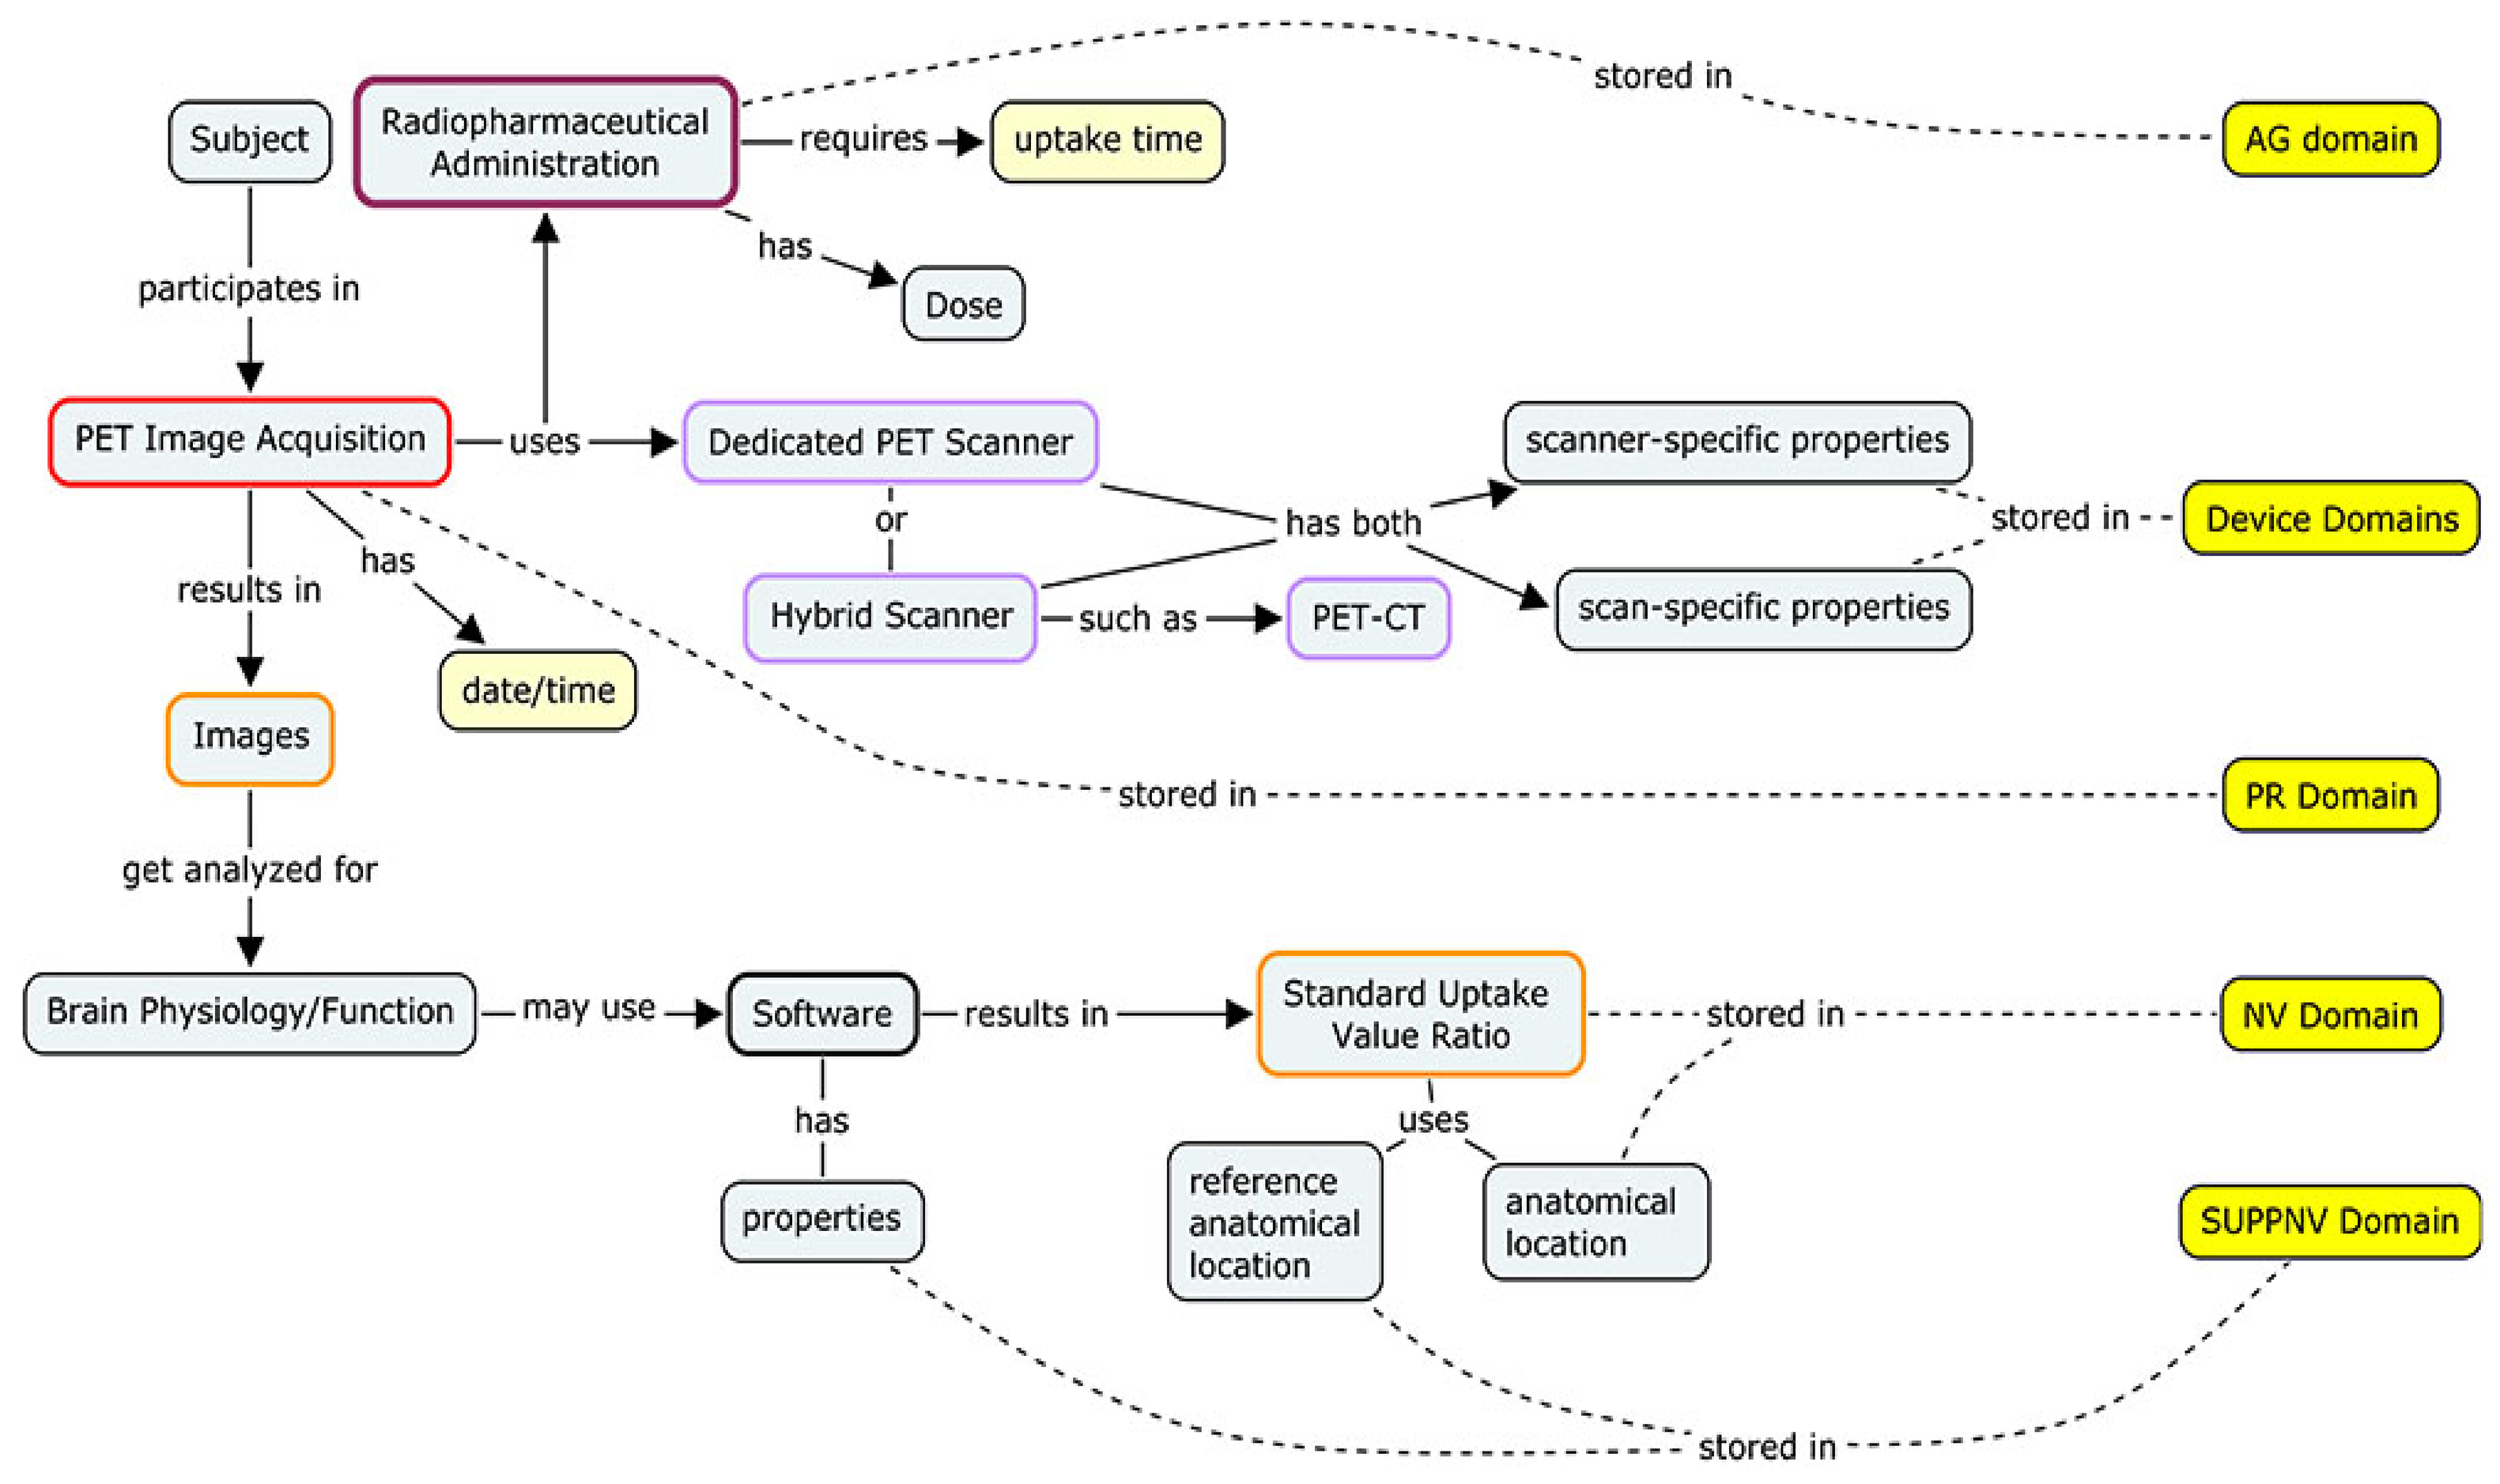
stored in
Subject
Radiopharmaceutical Administration
requires
uptake time
AG domain
has
Dose
participates in
PET Image Acquisition
uses
Dedicated PET Scanner
scanner-specific properties
or
has both
stored in
Device Domains
has
Hybrid Scanner
such as
PET-CT
scan-specific properties
results in
date/time
Images
stored in
PR Domain
get analyzed for
Brain Physiology/Function
may use
Software
results in
Standard Uptake Value Ratio
stored in
NV Domain
has
uses
properties
reference anatomical location
anatomical location
SUPPNV Domain
stored in

Panel B: Concept map of PET and PET-CT imaging

Figure 1: Concept modeling involves iterative discussions between clinical subject-matter experts and data modeling experts to parse the various concepts and relevant qualifiers that describe the pertinent information and data generated within a given research topic. The resulting "concept maps" (examples shown in panels A and B) are the first stage in the development of data models that describe how individual data elements relate to each other so that the resulting data model accounts for and preserves these relationships. The color-coding on the perimeter of grey boxes defines observation classes within the CDISC Bioinformatics Research Information Domain Grid (BRIDG) model. Yellow boxes correspond to the CDISC SDTM domain (dataset) where the concepts described reside.

Conclusions

The use of consensus data standards maximizes efficiency in regulatory review and facilitates analyses across diverse studies. CDISC standards allow for integrating and pooling data across various stakeholders' systems in a platform-independent manner. Implementation of CDISC standards, particularly in the biomarkers arena, promises to facilitate improved efficiencies and harmonization in clinical trials.

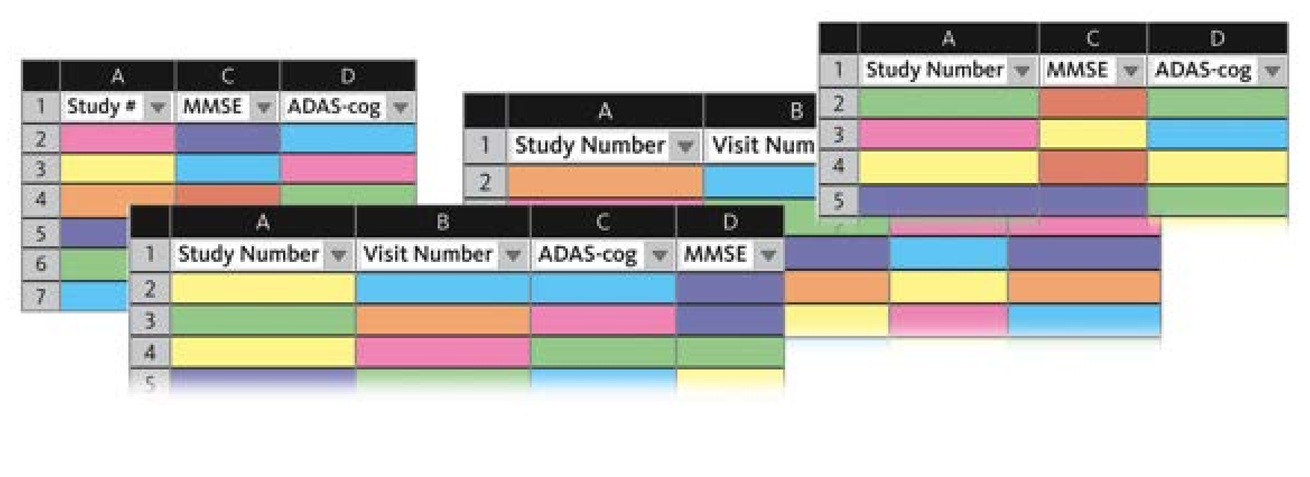
Study Number
Visit Number
MMSE
ADAS-cog

Unstandardized Data

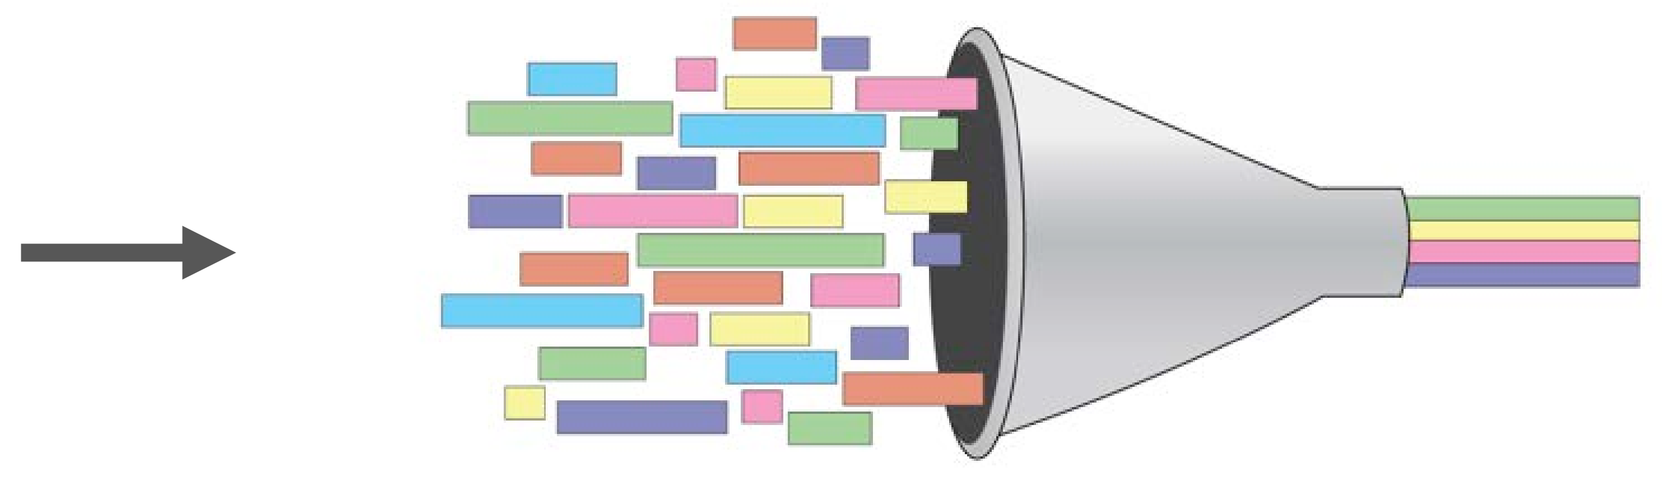

CDISC Data Standards

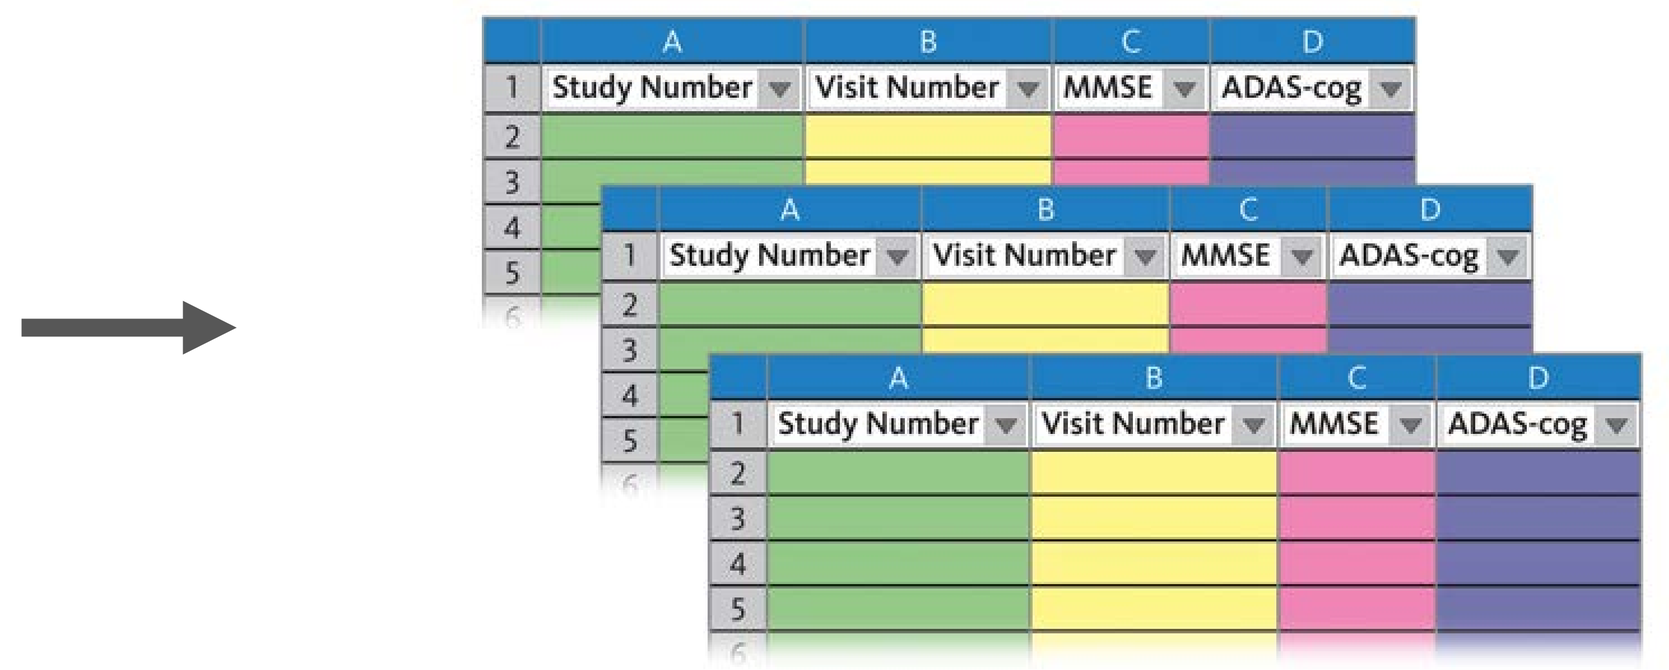
Study Number
Visit Number
MMSE
ADAS-cog

Standardized Datasets Facilitate Review & Analysis

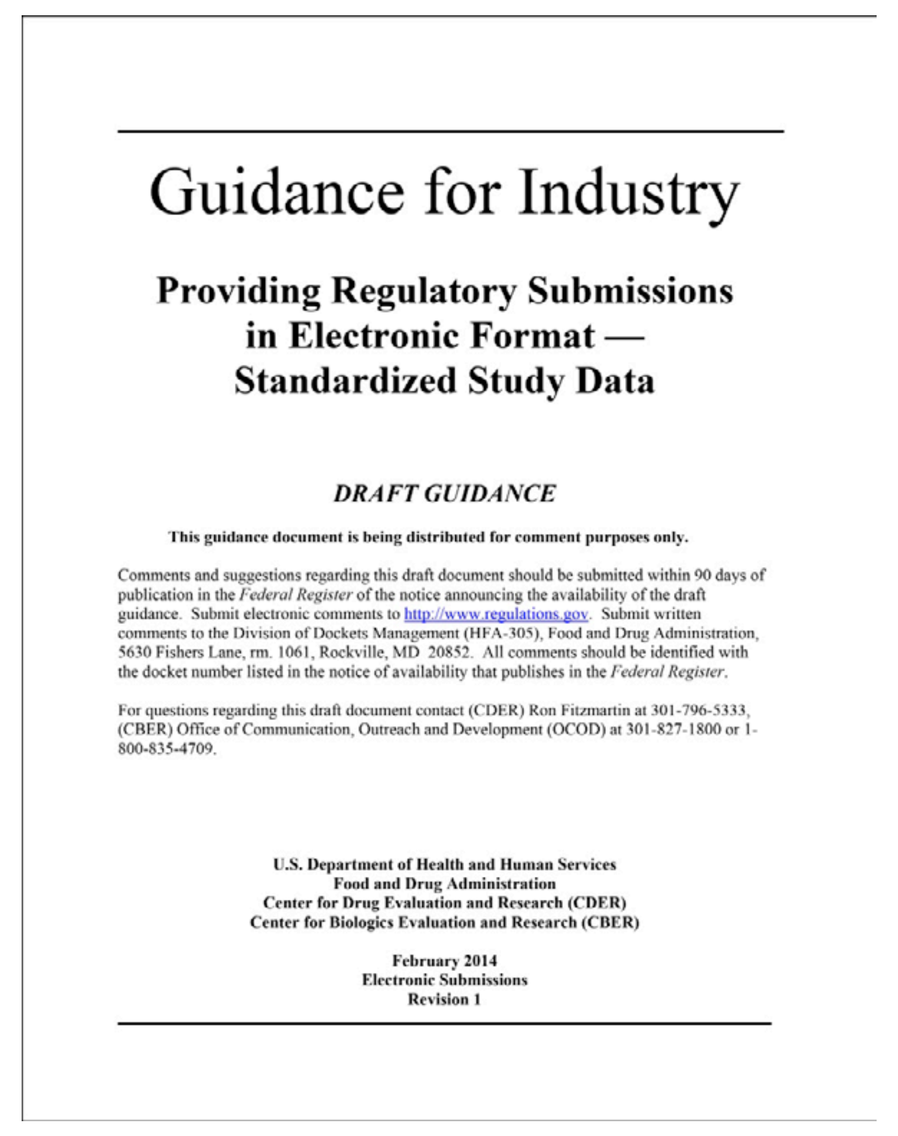
Guidance for Industry

Providing Regulatory Submissions in Electronic Format — Standardized Study Data

DRAFT GUIDANCE

U.S. Department of Health and Human Services
Food and Drug Administration
Center for Drug Evaluation and Research (CDER)
Center for Biologics Evaluation and Research (CBER)

February 2014
Electronic Submissions
Revision 1

Acknowledgments: The authors wish to thank Kewei Chen and Adam Fleisher of the Banner Alzheimer's Institute for their contributions to the PET imaging portion of the CDISC Alzheimer's User Guide.

CDISC CRITICAL PATH INSTITUTE CAMD CRITICAL PATH INSTITUTE